Neoadjuvant giredestrant (GDC-9545) + palbociclib (P) vs anastrozole (A) + P in postmenopausal women with estrogen receptor-positive, HER2-negative, untreated early breast cancer (ER+/HER2– eBC): final analysis of the randomized, open-label, international phase 2 coopERA BC study.

589

Peter A Fasching[1], Aditya Bardia[2], Vanesa Quiroga[3], Yeon H Park[4], Isabel Blancas[5], Jose L Alonso[6], Aleksandr Vasiliev[7], Hryhoriy Adamchuk[8], Marcelo Salgado[9], Denise A Yardley[10], Gonzalo Spera[11], Cloris Xue[12], Erika Ferreira[13], Tanja Badovinac Crnjevic[14], Pablo D Perez-Moreno[15], Vanesa López-Valverde[14], Jutta Steinseifer[14], Tharu M Fernando[15], Heather M Moore[15], Sara A Hurvitz[16]

[1]University Hospital Erlangen, Comprehensive Cancer Center (CCC) Erlangen-EMN, Friedrich-Alexander University Erlangen-Nürnberg, Erlangen, Germany; [2]Massachusetts General Hospital, Harvard Medical School, Boston, MA; [3]GEICAM Spanish Breast Cancer Group, San Sebastián de los Reyes, Madrid, Spain; [4]Samsung Medical Center, Seoul, Korea; [5]Hospital Universitario Clínico San Cecilio, Granada, Spain; [6]Hospital Clínico Universitario Virgen de la Arrixaca, El Palmar, Spain; [7]NSHI Road Clinical Hospital of JSC Russian Railways, Saint Petersburg, Russia; [8]Communal enterprise "Kryvyi Rih Oncology Dispensary", Kryvyi Rih, Ukraine; [9]Hospital dos Servidores do Estado de Pernambuco, Recife, Brazil; [10]Tennessee Oncology and Sarah Cannon Research Institute, Nashville, TN; [11]Translational Research in Oncology (TRIO), Montevideo, Uruguay; [12]F. Hoffmann-La Roche Ltd, Toronto, ON, Canada; [13]Roche Products Limited, Welwyn Garden City, UK; [14]F. Hoffmann-La Roche Ltd, Basel, Switzerland; [15]Genentech, Inc., South San Francisco, CA; [16]University of California, Los Angeles/Jonsson Comprehensive Cancer, Los Angeles, CA

BACKGROUND

- Endocrine therapy (ET), the therapeutic mainstay for estrogen receptor-positive breast cancer (ER+ BC), targets ER activation and/or estrogen synthesis; however, many of these patients experience treatment resistance or disease relapse.[1,2]
- Giredestrant is a highly potent, nonsteroidal, oral, selective ER antagonist and degrader (SERD) that achieves robust ER occupancy, is well tolerated, and has previously shown encouraging antitumor activity as a monotherapy and in combination with the cyclin-dependent kinase 4/6 inhibitor palbociclib in metastatic BC (mBC).[3–7]
- coopERA BC (NCT04436744) is a phase 2 randomized trial in ER-positive, HER2-negative, untreated early BC (ER+/HER2– eBC) that was designed to test the hypothesis of whether giredestrant has a stronger antiproliferative effect (measured by Ki67) than anastrozole, both as monotherapy, after 2 weeks of treatment in a window-of-opportunity phase. As a secondary objective, the trial investigated changes in Ki67 and clinical efficacy after 16 weeks of neoadjuvant combination treatment with giredestrant plus palbociclib vs anastrozole plus palbociclib. The study met its primary efficacy endpoint, showing superior Ki67 suppression with giredestrant over anastrozole after all subjects had completed week 2 (Table 1), with safety data remaining consistent with the known safety profile.[8]
- Here, we report the final analysis after 16 weeks of neoadjuvant treatment.

METHODS

- The study design is shown in Figure 1.

Figure 1: Study design

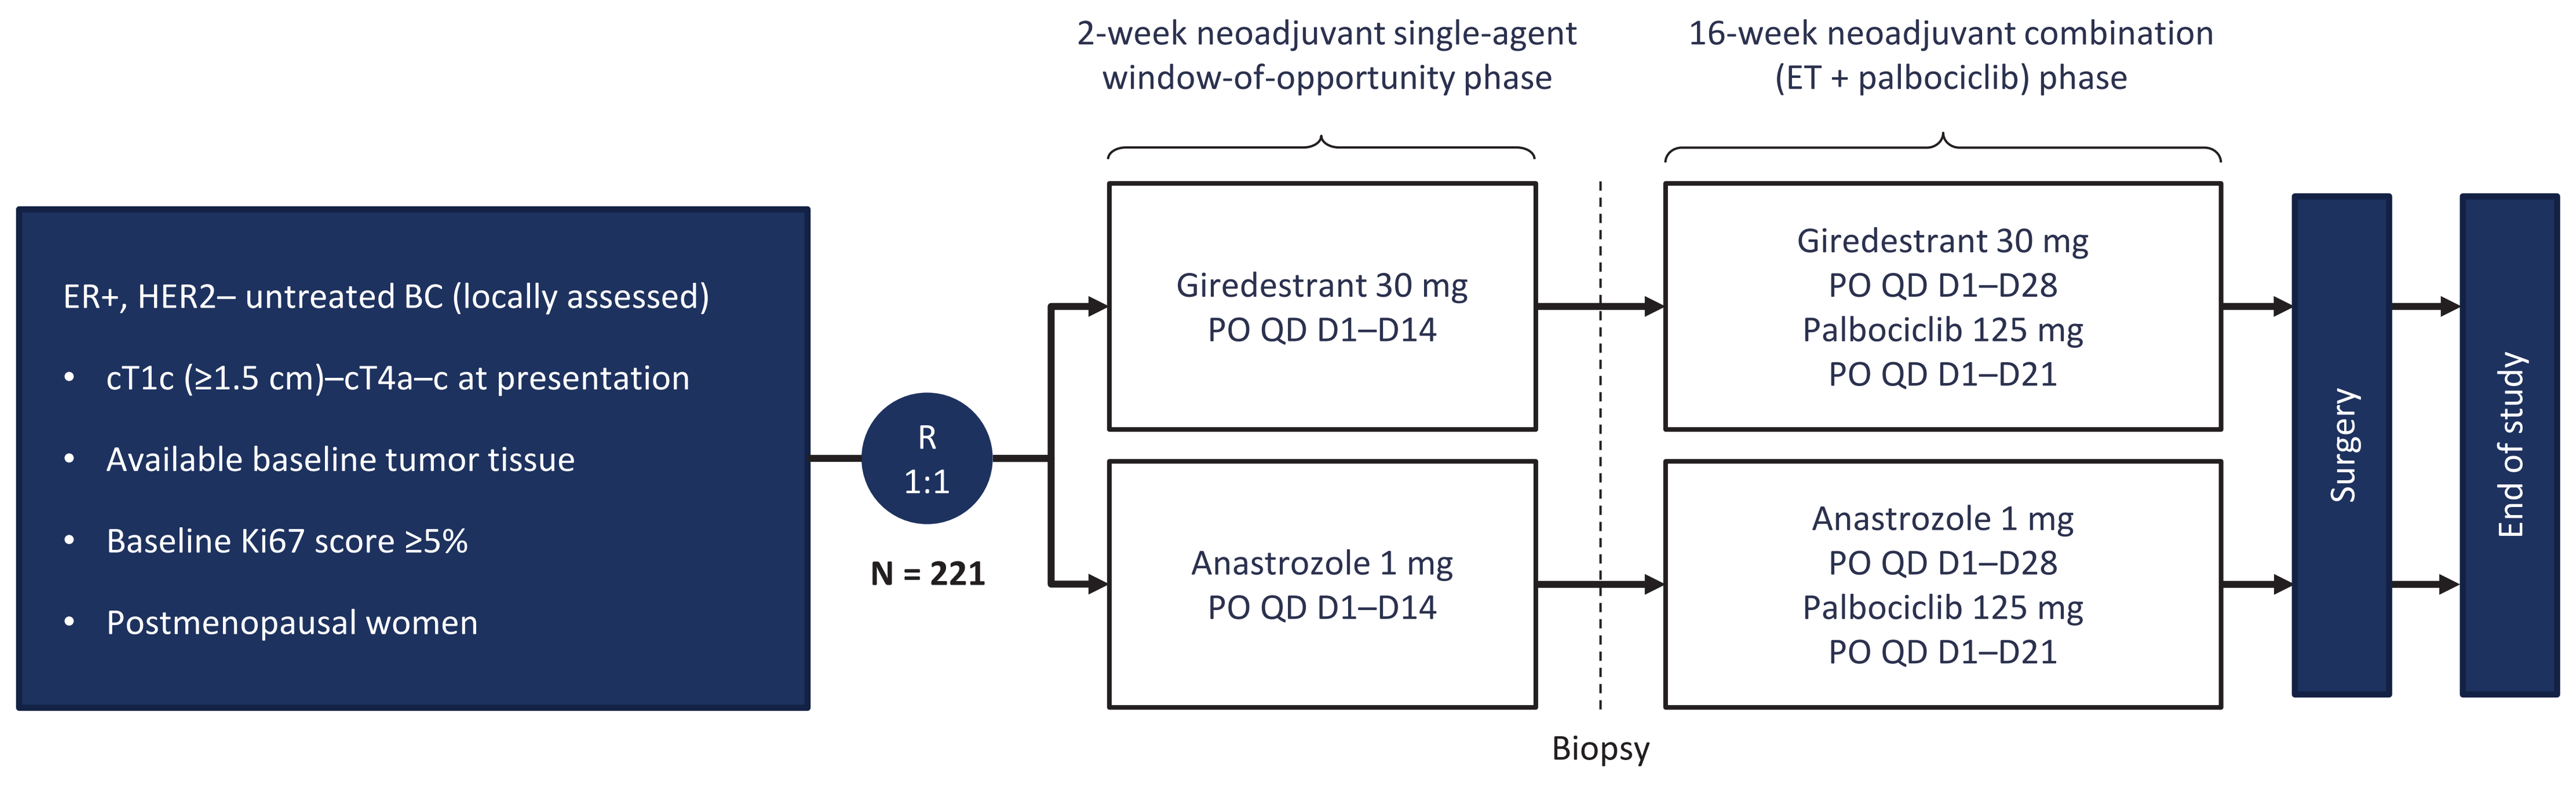
2-week neoadjuvant single-agent window-of-opportunity phase
16-week neoadjuvant combination (ET + palbociclib) phase
ER+, HER2– untreated BC (locally assessed)
• cT1c (≥1.5 cm)–cT4a–c at presentation
• Available baseline tumor tissue
• Baseline Ki67 score ≥5%
• Postmenopausal women
R 1:1
N = 221
Giredestrant 30 mg PO QD D1–D14
Anastrozole 1 mg PO QD D1–D14
Giredestrant 30 mg PO QD D1–D28 Palbociclib 125 mg PO QD D1–D21
Anastrozole 1 mg PO QD D1–D28 Palbociclib 125 mg PO QD D1–D21
Biopsy
Surgery
End of study

- Randomization was stratified by tumor size, baseline Ki67 score, and progesterone receptor status.
- Endpoints assessed here include Ki67 suppression from baseline to surgery, CCCA (Ki67 ≤2.7%) at surgery, ORR, pCR, and safety.

Adapted from Hurvitz SA, *et al*. 2021.[8]
BC, breast cancer; CCCA, complete cell cycle arrest; D, day; ER+, estrogen receptor-positive; ET, endocrine therapy; ORR, objective response rate; pCR, pathologic complete response; PO QD, oral daily; R, randomization.

RESULTS

Patients

- At final analysis (cutoff: November 24, 2021), 112 and 109 patients were randomized to the giredestrant and anastrozole arms, respectively.
- Median age was 62 years (both arms) and the percentages of patients with stage I/IIa disease were 60% in the giredestrant arm and 54% in the anastrozole arm.
- Median treatment duration was 128 days (range: 1–105) for giredestrant and 105 (range: 6–128) for palbociclib in the giredestrant arm, and 129 (range: 14–158) for anastrozole and 105 (range: 21–133) for palbociclib in the anastrozole arm.
- Median times from last exposure to surgery sample collection were 9 days (range: –19 to 36) and 10 days (range: –6 to 54) for the giredestrant and anastrozole arms, respectively.

Ki67 change from baseline to surgery

- Consistent with the primary analysis, giredestrant plus palbociclib showed a greater suppression of Ki67 at surgery compared with anastrozole plus palbociclib (Figures 2 and 3; Table 1).

Ki67 change at surgery by baseline high vs low Ki67

- Similar to the week 2 response, giredestrant in combination with palbociclib remained superior to the anastrozole combination in terms of Ki67 reduction at surgery in patients whose tumors had baseline Ki67 scores ≥20% (Table 2A).

Complete cell cycle arrest (CCCA) at surgery

- Greater CCCA rates of giredestrant vs anastrozole in combination with palbociclib were maintained at surgery (Table 2B).
- Superior CCCA was observed in patients with baseline Ki67 ≥20% or <20% (Table 2B).

Activity

- Objective response rate (ORR) was similar between the two arms (giredestrant + palbociclib: 50% [95% confidence interval (CI): 40%, 60%]; anastrozole + palbociclib: 49% [95% CI: 39%, 59%]).
- Pathologic complete response (pCR) rates were 4.5% (95% CI: 1.5%, 10.1%) and 4.6% (1.5%, 10.5%), respectively.

Safety

- ET-related adverse events (AEs) were non-serious and occurred at similar rates between the two arms (Table 3).
- ET-related Grade ≥3 AE rates were also similar at 6% each. The most common was neutropenia (4% per arm).
- Interruption/withdrawal of ET due to AEs was low and similar for both arms (Table 3).

Figure 2: Relative reduction in Ki67 at week 2[8] and at surgery

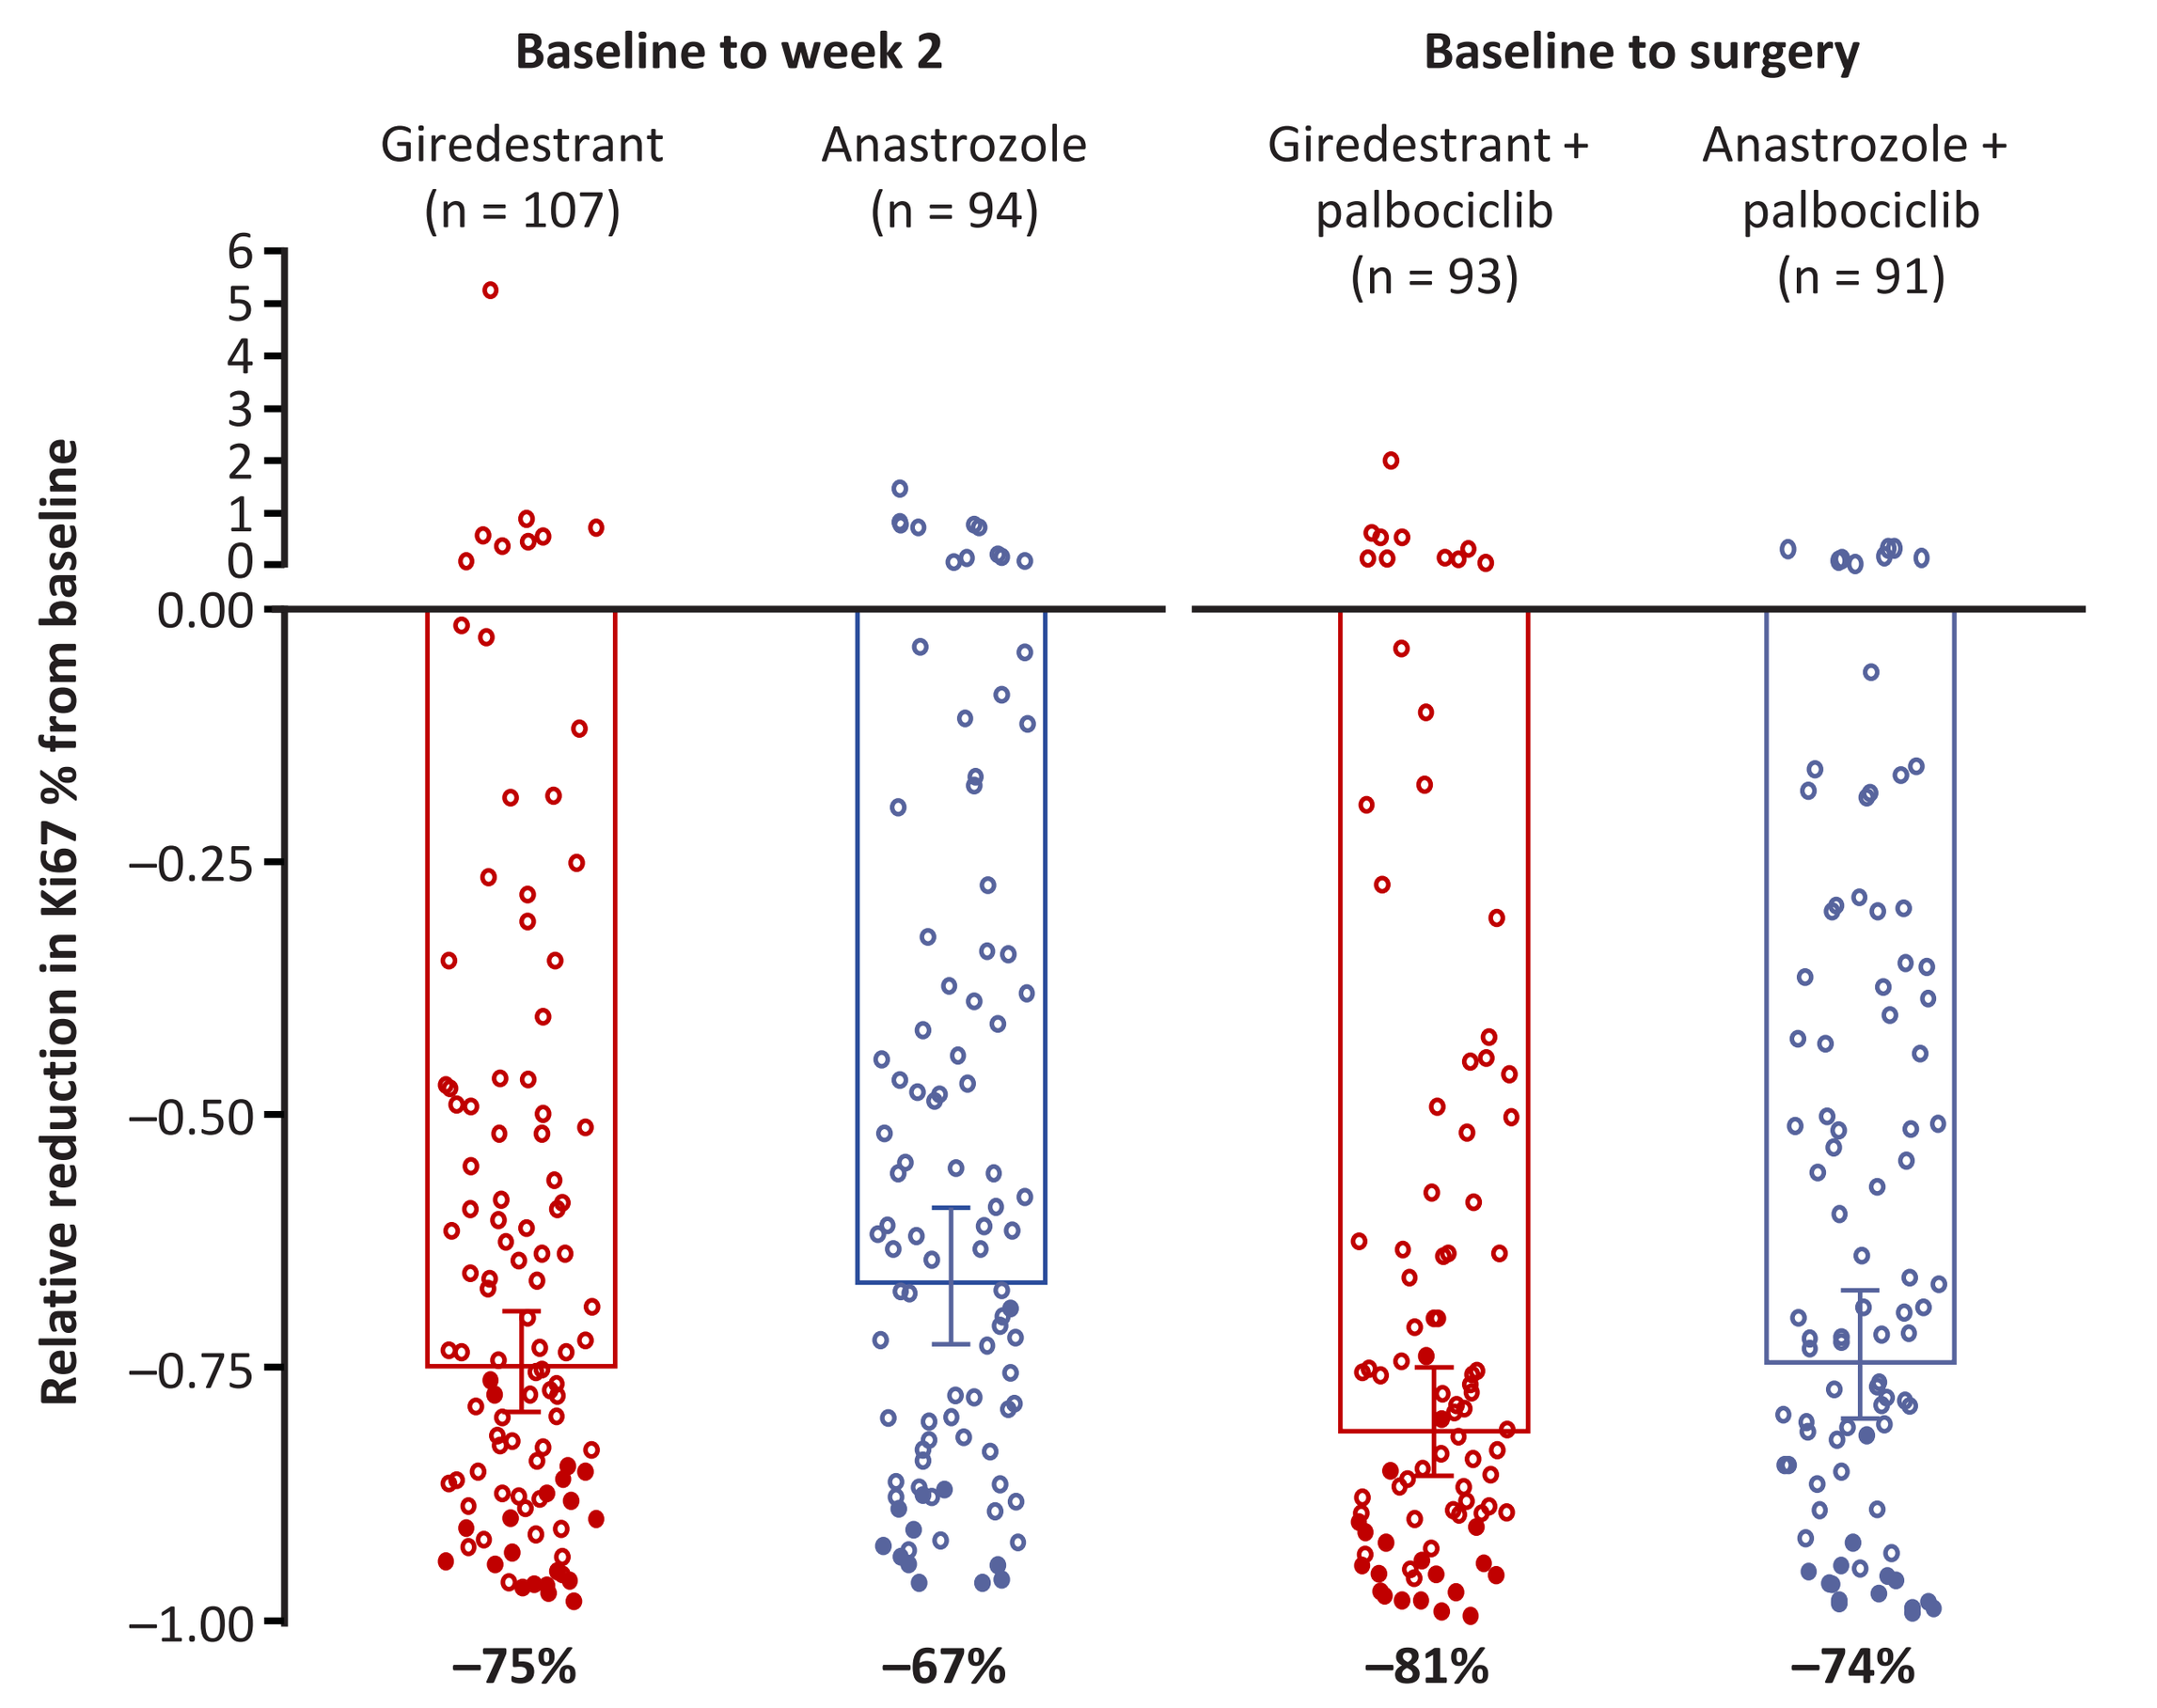
Baseline to week 2
Baseline to surgery
Giredestrant (n = 107)
Anastrozole (n = 94)
Giredestrant + palbociclib (n = 93)
Anastrozole + palbociclib (n = 91)
Relative reduction in Ki67 % from baseline
6
5
4
3
2
1
0
0.00
–0.25
–0.50
–0.75
–1.00
–75%
–67%
–81%
–74%

Baseline is the patient's last observation prior to initiation of study drug. Bars represent the geometric mean of % relative reduction based on the relative change of Ki67 at week 2 or surgery compared with baseline along with 95% confidence intervals. Solid circles represent Ki67 ≤2.7% (complete cell cycle arrest); open circles, Ki67 >2.7%.

Table 1: Relative reduction in Ki67 % from baseline to week 2[8] and to surgery

	Giredestrant + palbociclib	Anastrozole + palbociclib
To week 2	**n = 107**	**n = 94**
	Proportional change from baseline	
Geometric mean (95% CI)	0.25 (0.20, 0.30)	0.33 (0.27, 0.41)
Minimum–maximum	0.03–6.21	0.03–2.40
	% relative reduction from baseline	
Geometric mean (95% CI)	–75% (–80%, –70%)	–67% (–73%, –59%)
To surgery	**n = 93**	**n = 91**
	Proportional change from baseline	
Geometric mean (95% CI)	0.19 (0.14, 0.25)	0.26 (0.20, 0.33)
Minimum–maximum	0.00–2.96	0.01–1.35
	% relative reduction from baseline	
Geometric mean (95% CI)	–81% (–86%, –75%)	–74% (–80%, –67%)

CI, confidence interval.

Figure 3: Patient Ki67 scores by visit

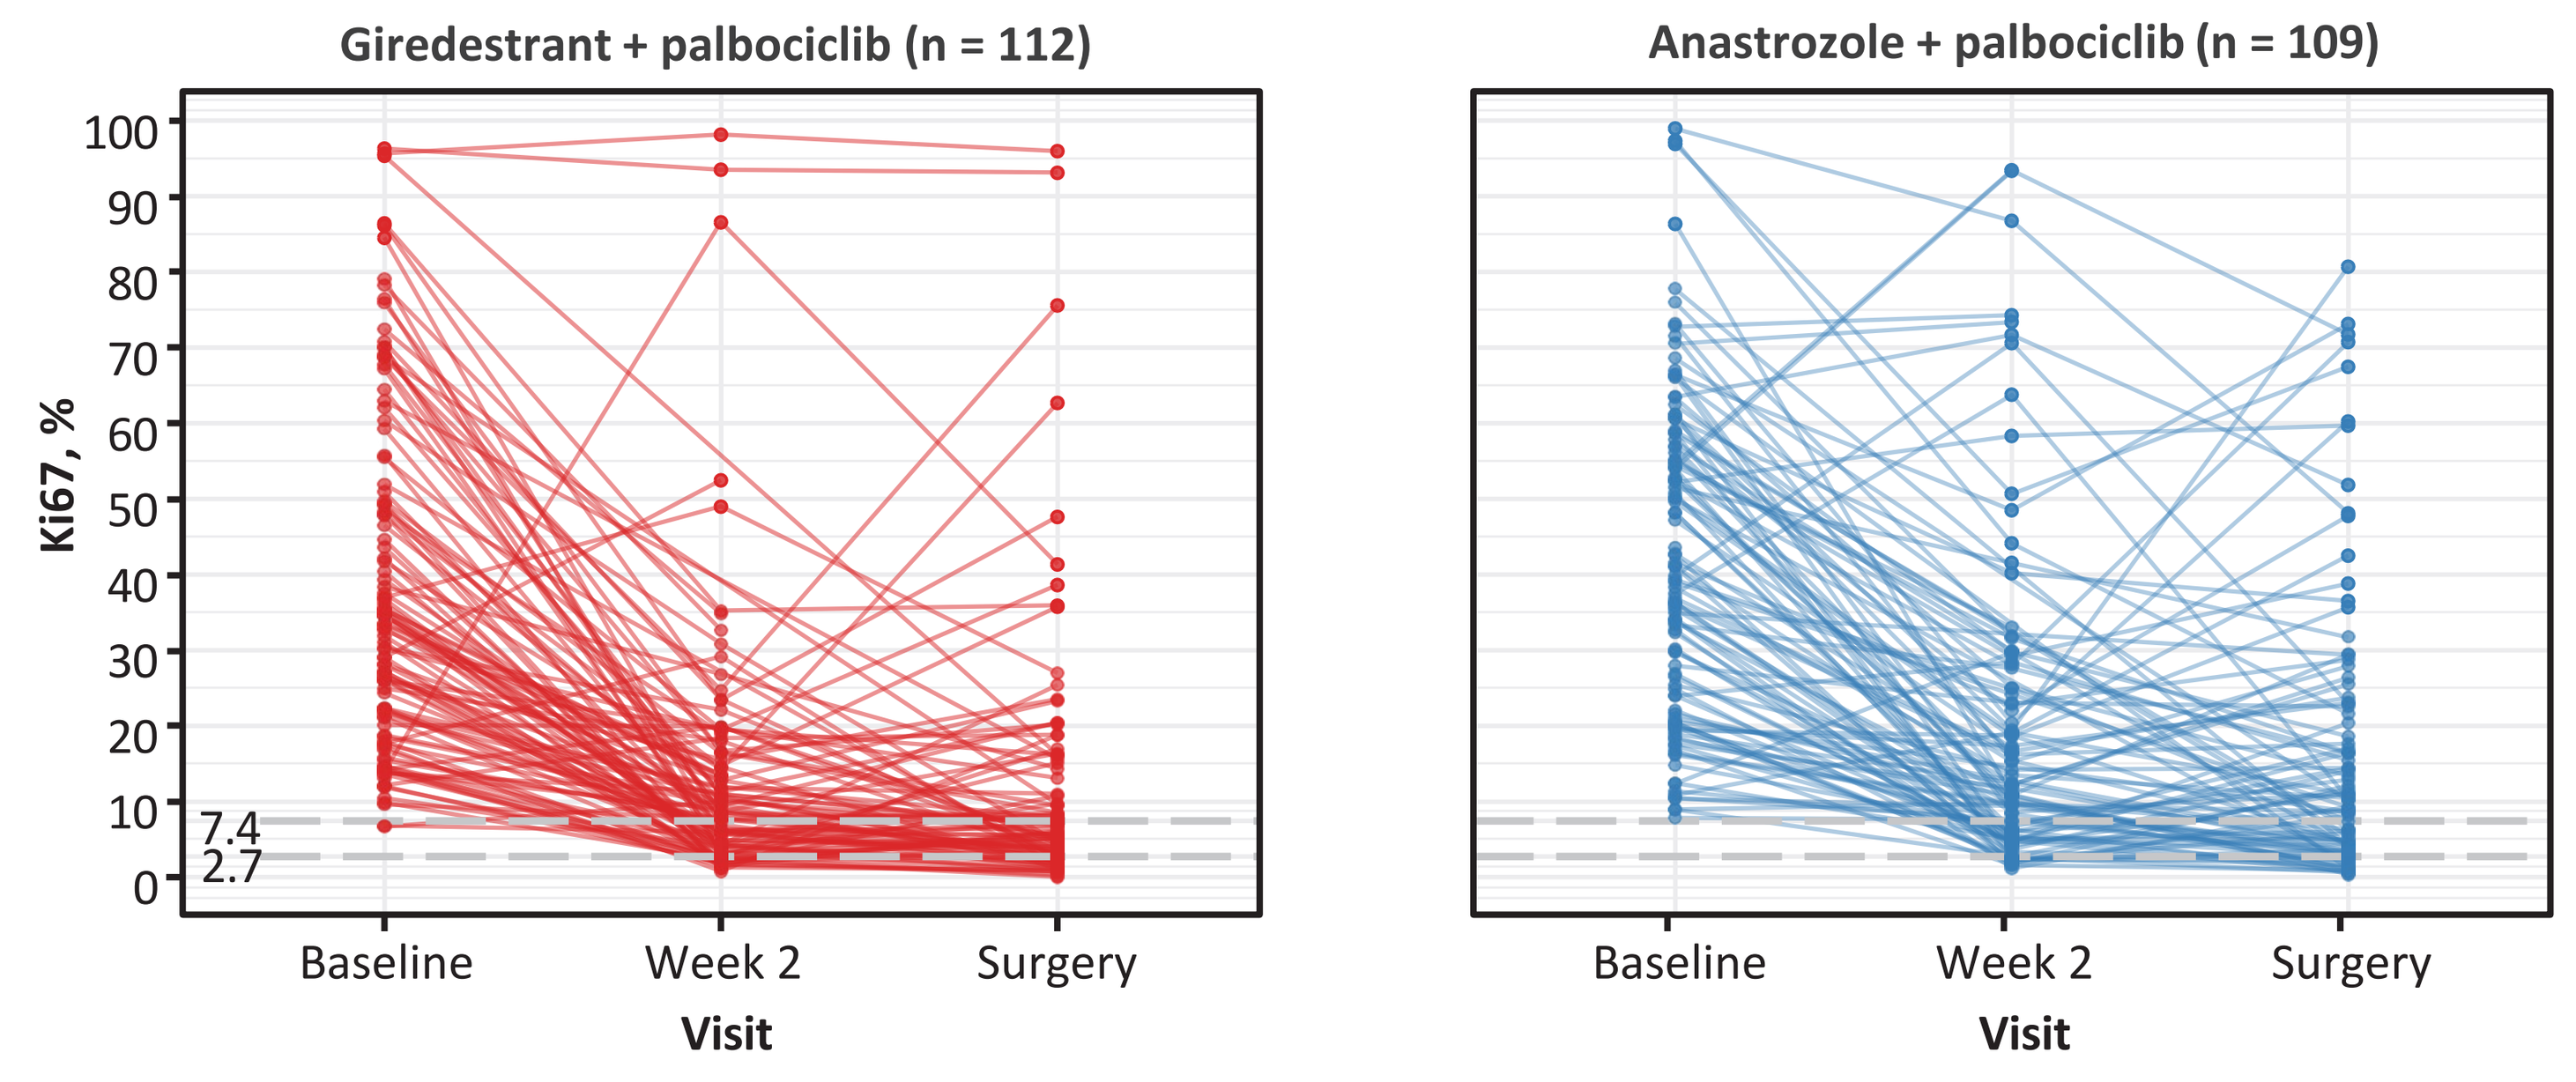
Giredestrant + palbociclib (n = 112)
Anastrozole + palbociclib (n = 109)
Ki67, %
100
90
80
70
60
50
40
30
20
10
7.4
2.7
0
Baseline
Week 2
Surgery
Visit

Baseline is the patient's last observation prior to initiation of study drug. Horizontal lines labeled 2.7% and 7.4% reflect Ki67 values of 2.7499% and 7.4499%, respectively.

Table 2: A) Ki67 response at week 2[8] and at surgery by baseline high vs low Ki67 B) CCCA at week 2[8] and at surgery

A	Baseline high Ki67 ≥20%		Baseline low Ki67 <20%	
	Giredestrant + palbociclib	Anastrozole + palbociclib	Giredestrant + palbociclib	Anastrozole + palbociclib
Week 2	**n = 80**	**n = 77**	**n = 27**	**n = 17**
Geometric mean of % relative reduction (95% CI)	–79% (–83%, –74%)	–70% (–76%, –63%)	–59% (–73%, –37%)	–43% (–63%, –12%)
CCCA, n (%)	12 (15%)	9 (12%)	9 (33%)	3 (18%)
Surgery	**n = 68**	**n = 71**	**n = 25**	**n = 20**
Geometric mean of % relative reduction (95% CI)	–83% (–87%, –77%)	–75% (–81%, –67%)	–74% (–87%, –49%)	–73% (–84%, –55%)
CCCA, n (%)	13 (19%)	11 (15%)	9 (36%)	4 (20%)

B	Giredestrant + palbociclib	Anastrozole + palbociclib
Week 2	**n = 107**	**n = 94**
CCCA, n (%)	21 (20%)	12 (13%)
Difference between arms, % (95% CI)	7% (–4%, 18%)	
Surgery	**n = 93**	**n = 91**
CCCA, n (%)	22 (24%)	15 (16%)
Difference between arms, % (95% CI)	7% (–5%, 20%)	

CCCA, complete cell cycle arrest; CI, confidence interval.

Table 3: Safety overview

Patients, n (%)	Giredestrant + palbociclib (n = 112)	Anastrozole + palbociclib (n = 109)
Total number of patients with ≥1 AE	104 (93%)	98 (90%)
Grade 3–4 AEs	49 (44%)	47 (43%)
Grade 5 AEs	1 (1%)*	0
AEs leading to ET interruption	5 (5%)	5 (5%)
AEs leading to ET withdrawal	2 (2%)	1 (1%)
ET-related AEs	54 (48%)	52 (48%)
Serious AEs	5 (5%)	2 (2%)
Hip fracture	1 (1%)	0
Procedural pain	0	1 (1%)
Uterine perforation	1 (1%)	0
Myocardial infarction	1 (1%)	0
Pyrexia	1 (1%)	0
COVID-19	1 (1%)	0
Hypoxia	0	1 (1%)

Patients, n (%)	Giredestrant + palbociclib (n = 112)	Anastrozole + palbociclib (n = 109)
AE with ≥5% difference between treatment arms		
Fatigue	10 (9%)	18 (17%)
Grade 3–4	0	0
Anemia	12 (11%)	6 (6%)
Grade 3–4	0	0
Mucosal inflammation	9 (8%)	3 (3%)
Grade 3–4	0	0
Arthralgia	12 (11%)	21 (19%)
Grade 3–4	0	0
Diarrhea	8 (7%)	18 (17%)
Grade 3–4	0	0
Alanine aminotransferase increased	1 (1%)	9 (8%)
Grade 3–4	1 (1%)	3 (3%)

* Myocardial infarction. AE, adverse event; ET, endocrine therapy. All serious AEs were assessed as unrelated to study treatment.

CONCLUSIONS

- **In this final analysis of coopERA BC, the greater suppression of Ki67 with giredestrant vs anastrozole observed at week 2 in the primary analysis[8] was maintained at surgery following the addition of palbociclib.**
- **CCCA also remained higher with giredestrant vs anastrozole at surgery, as it was at week 2.[8]**
- **ORR was similar in both arms. This was expected given the primarily cytostatic action of ET and the inherent biology of ER+ tumors, and was in line with experience from other trials in this setting.[9–15]**
- **pCR rates were also in line with previous trials in the ER+ setting.[14,16] pCR is a rare event with ET and chemotherapy, and is not correlated with long-term outcomes.**
- **Safety data remained consistent with the known safety profile of giredestrant.[3,5,17,18]**
- **coopERA BC is the first randomized study to show superior antiproliferative activity of an oral SERD (giredestrant) over an aromatase inhibitor (anastrozole) in ER+/HER2– eBC.**
- **The results from coopERA BC support investigating giredestrant vs ET in the adjuvant setting.**

REFERENCES

1. Reinert T & Barrios CH. *Ther Adv Med Oncol* 2015; **7**:304–320; 2. Osborne CK & Schiff R. *Annu Rev Med* 2011; **62**:233–247; 3. Jhaveri KL, *et al*. ASCO 2021; 1017; 4. Liang J, *et al*. *J Med Chem* 2021; **64**:11841–11856; 5. Moore HM, *et al*. ASCO 2021; 577; 6. Jhaveri KL, *et al*. SABCS 2020; PD7-05; 7. Metcalfe C, *et al*. SABCS 2019; P5-04-7; 8. Hurvitz SA, *et al*. SABCS 2021; PD-13-06; 9. Johnston S, *et al*. *J Clin Oncol* 2019; **37**:178–189; 10. Dowsett M, *et al*. *Clin Cancer Res* 2005; **11**:951s–958s; 11. Smith I, *et al*. *Lancet Oncol* 2020; **21**:1443–1454; 12. Martin M, *et al*. SABCS 2017; PD5-01; 13. Khan QJ, *et al*. ASCO 2020; 505; 14. Ma CX, *et al*. *Clin Cancer Res* 2017; **23**:4055–4065; 15. Ellis MJ, *et al*. *J Natl Cancer Inst* 2008; **100**:1380–1388; 16. Brandão M & Ignatiadis M. *Ann Oncol* 2018; **29**:2274–2278; 17. Lim E, *et al*. ASCO 2020; 1023; 18. Neilan T, *et al*. SABCS 2021; P5-18-07.

ACKNOWLEDGMENTS

We thank the patients and their families who took part in the study, as well as the staff, research coordinators, and investigators at each participating institution. Research support for third-party writing assistance for this poster, furnished by Laura Pérez-Pachón, PhD, MSc, of Health Interactions, was provided by F. Hoffmann-La Roche Ltd.

CONFLICTS OF INTEREST

PAF reports honoraria from Roche, Novartis, Pfizer, Daiichi Sankyo, Eisai, Merck Sharp & Dohme, AstraZeneca, Hexal, Lilly, Pierre Fabre, Seagen, Agendia, Gilead, Cepheid, and BioNTech; consulting or advisory role for Roche, Novartis, Pfizer, Daiichi Sankyo, Eisai, Merck Sharp & Dohme, AstraZeneca, Hexal, Pierre Fabre, Seagen, Agendia, Lilly, Gilead (self); research funding from Novartis, BioNTech, and Cepheid (institution).
Please refer to the abstract for all author conflicts of interest. All authors have received research support in the form of third-party writing assistance for this poster from F. Hoffmann-La Roche Ltd.
This study was sponsored by F. Hoffmann-La Roche Ltd.

This presentation is the intellectual property of the authors. Contact Peter.Fasching@uk-erlangen.de for permission to reprint and/or distribute.